Abstract ID: 2550

NCT03522298

Phase 2a study to evaluate the safety, pharmacokinetics, and clinical activity of the PI3K / mTOR inhibitor GDC-0084 given to glioblastoma (GBM) patients with unmethylated MGMT promotor status

P.Y. Wen[1], J. de Groot[2], J.D. Battiste[3], S.A. Goldlust[4], J.S. Garner[5], J. Simpson[5], J. Kijlstra[6], A. Olivero[7], T. Cloughesy[8]

[1]Center for Neuro-Oncology, Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, United States; [2]Department of Neuro-Oncology, Division of Cancer Medicine, The University of Texas MD Anderson Cancer Center, Houston, TX, United States; [3]Stephenson Cancer Center, University of Oklahoma, Oklahoma City, OK, United States; [4]John Theurer Cancer Center, Hackensack University Medical Center, Hackensack, NJ, United States; [5]Kazia Therapeutics Limited, Sydney, Australia; [6]Covance Inc., Princeton, NJ, United States; [7]Genentech Inc, South San Francisco, CA, United States; [8]Department of Neurology, Ronald Reagan UCLA Medical Center, University of California, Los Angeles, CA, United States

BACKGROUND

- **GDC-0084** (paxalisib) is a potent, oral, selective, brain-penetrant inhibitor of class I phosphoinositide 3-kinase and mammalian target of rapamycin[1,2].
- The **PI3K pathway** is upregulated in ~85% of GBM cases per the Cancer Genome Atlas[3], and GDC-0084 has shown efficacy in a range of preclinical models
- A **phase I study** (NCT01547546) investigated GDC-0084 given once-daily as an oral (PO) dose in 47 patients with recurrent high-grade gliomas. The maximum tolerated dose (MTD) was 45mg once daily[4].

OBJECTIVES

The current **phase IIa study** (NCT03522298) aims to explore the safety, tolerability, and clinical activity of GDC-0084 in patients with **newly-diagnosed GBM** and **unmethylated MGMT promotor** status, following surgical resection and chemoradiotherapy.

METHODS

This is an open-label, single-arm, multicenter study in two parts, as shown in Figure 1.

- **Stage 1** – a dose escalation cohort to establish the MTD in newly-diagnosed unmethylated patients
- **Stage 2** – a dose expansion cohort to seek preliminary evidence of clinical activity in newly-diagnosed patients

Figure 1: Study Design for Phase II study of GDC-0084

Stage 1: Dose Escalation

Standard "3+3" design:
- determine MTD in newly-diagnosed patients
- further define safety, tolerability and PK

9 patients enrolled

Stage 2: Expansion Cohort

Two-arm, open-label design:
- assess single agent activity of GDC-0084
- explore effect of fed vs. fasting state on PK

21 patients enrolled

SAFETY AND TOLERABILITY

Results from Stage 1 of the study (MTD determination) are reported

Demographics (*n*=9)

Figure 2: Key Demographic Parameters in Stage 1

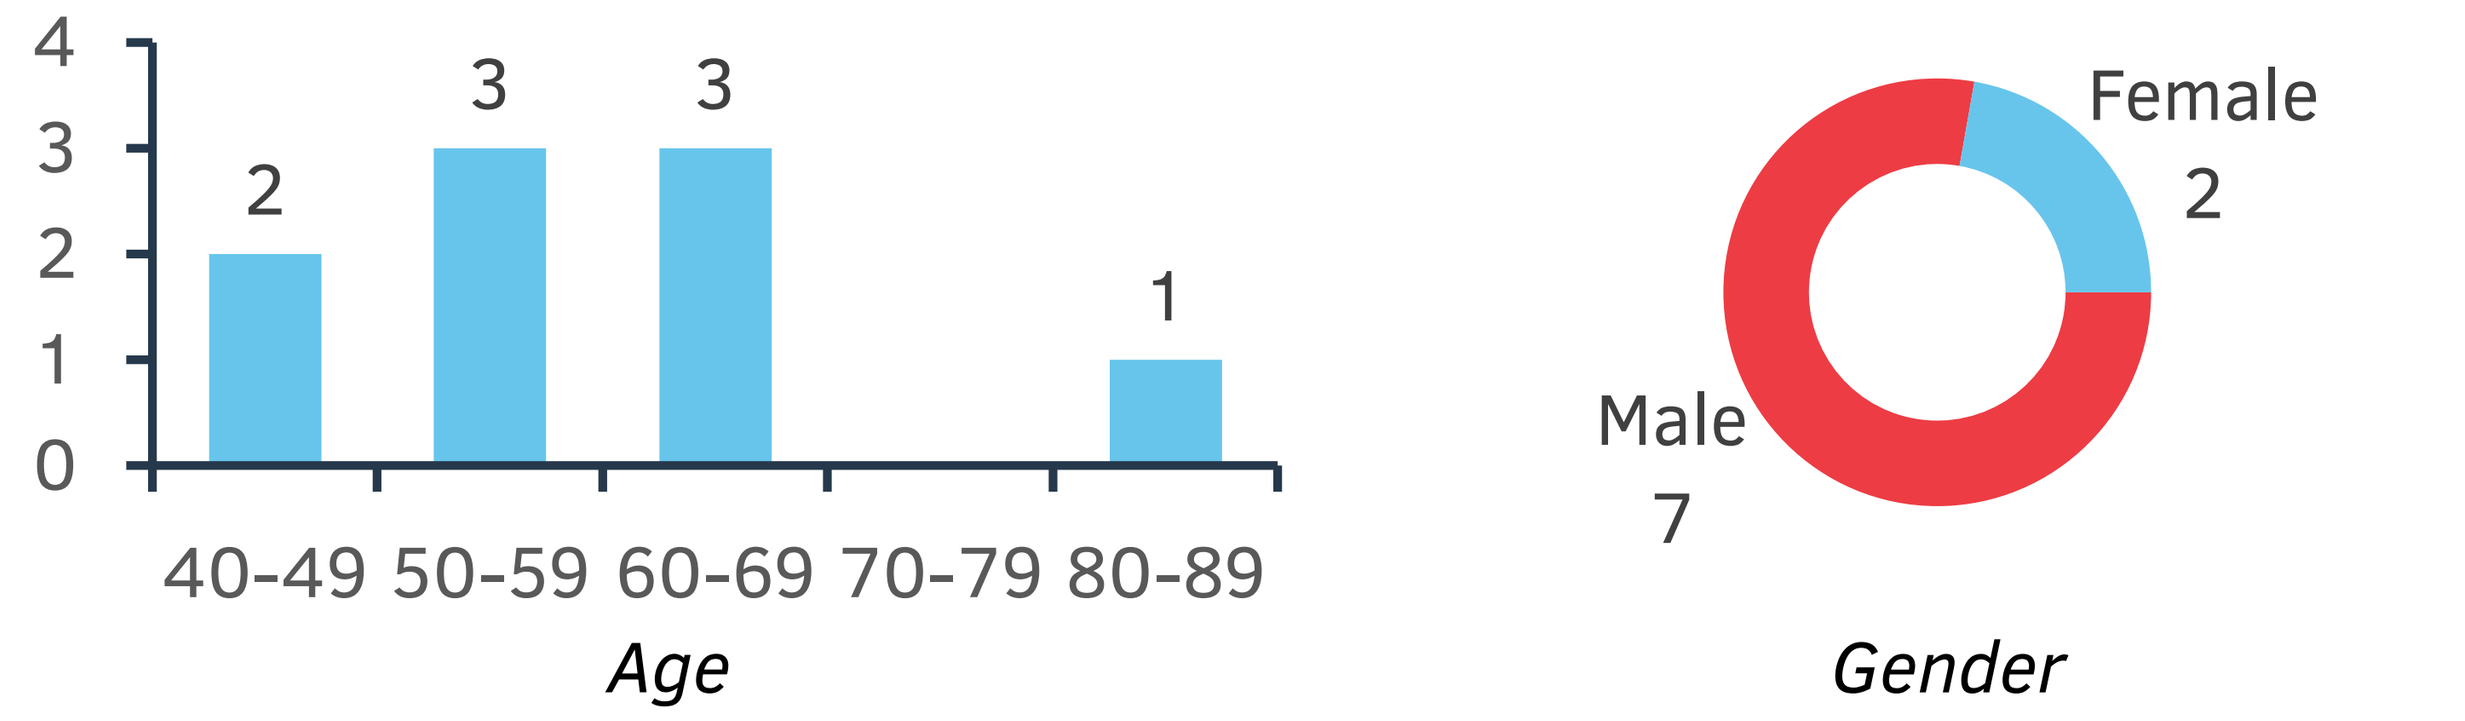
4
3
2
1
0
2
3
3
1
40-49 50-59 60-69 70-79 80-89
Age
Female
2
Male
7
Gender

Dose-Limiting Toxicities

Cohort	*n*	# DLTs	DLTs observed
60mg po od	3	0	
75mg po od	6	2	hyperglycemia; mucositis

Adverse Events

Table 1: Patients Experiencing Treatment-Emergent AEs 'Related' or 'Possibly Related' to GDC-0084 (only terms with >1 patient)

	Gr 1	Gr 2	Gr 3	Gr 4	Total
SKIN RASH	2		4		6
ORAL MUCOSITIS	2		2		4
HYPERGLYCEMIA	2	2	1	2	7
FATIGUE	1	4			5
DECR. LYMPHOCYTES	2	1			3
DECR. NEUTROPHILS	1	1			2
DECR. PLATELETS	3	1			4
DIARRHEA	2				2
NAUSEA	2	2			4
VOMITING	2	1			3
WEIGHT LOSS	3				3
ANOREXIA		2			2
HYPERTRIGLYCERIDEMIA	1	2			3
HYPERCHOLESTEROLEMIA	2				2
HYPOPHOSPHATEMIA		1	1		2

EFFICACY

- For the eight evaluable patients in Stage 1, a median progression-free survival (PFS) of **8.4 months** was determined (Figure 3).
- One patient remains progression-free and on treatment nineteen months after diagnosis.
- A median overall survival (OS) of **17.7 months** was determined (Figure 4). At the analysis cut-off, three of eight patients (38%) remained alive.

Figure 3: Kaplan-Meier Curve of Progression-Free Survival (PFS)

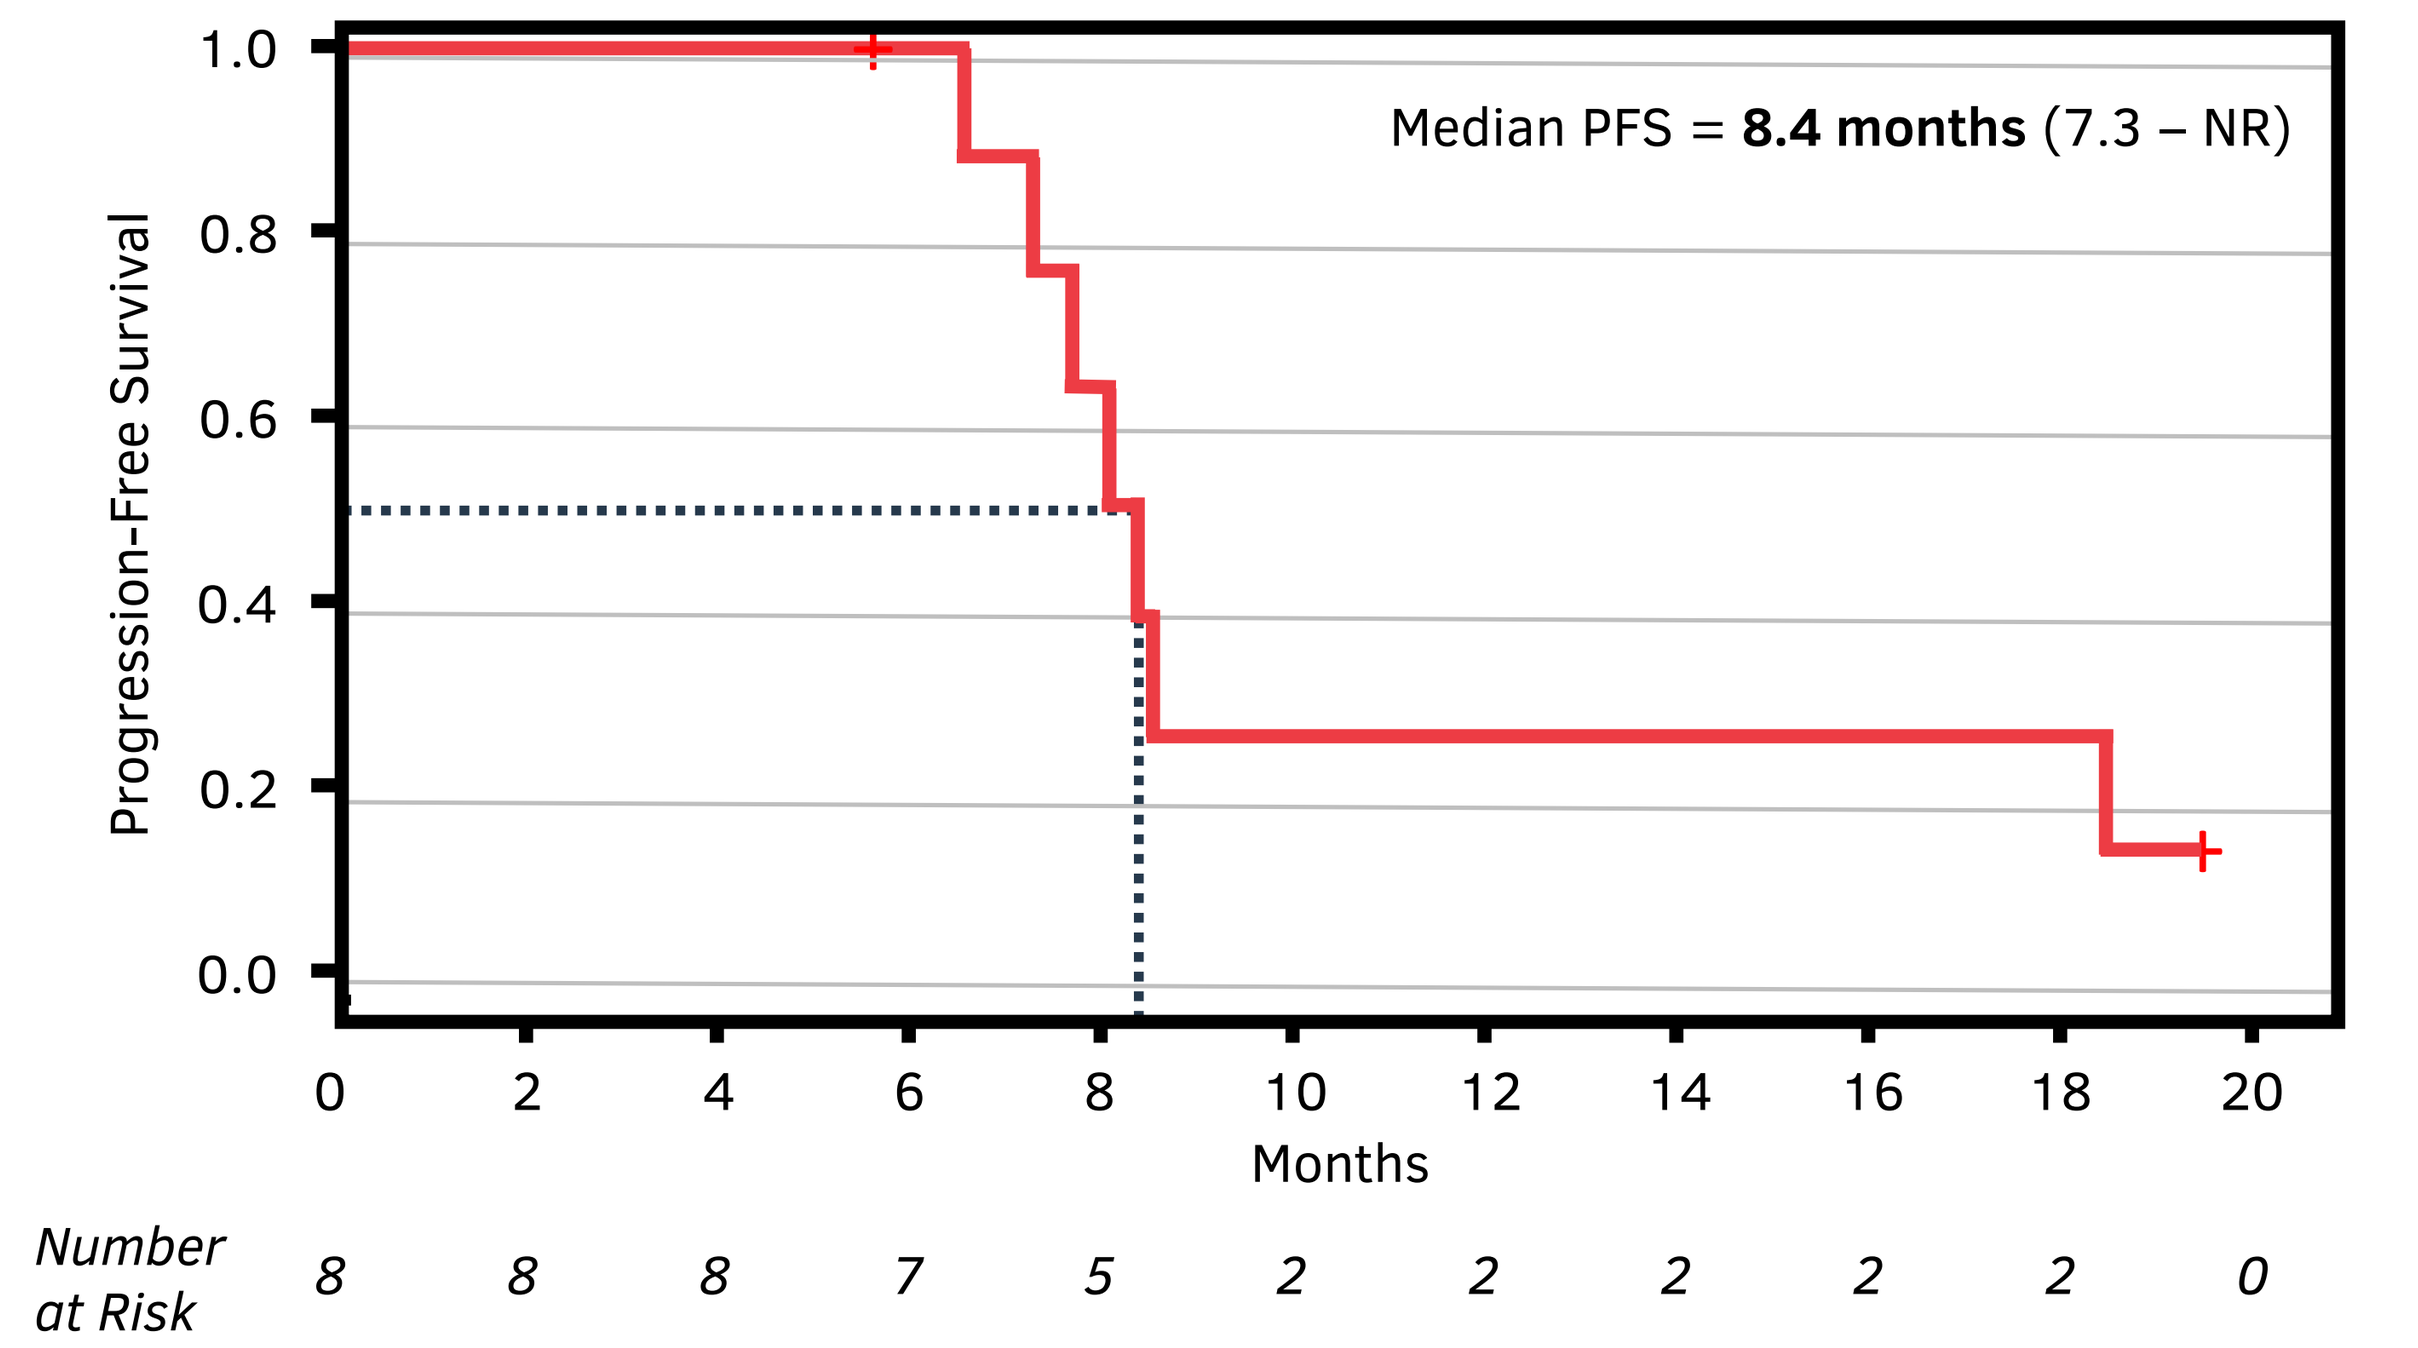
Median PFS = 8.4 months (7.3 – NR)
Progression-Free Survival
1.0
0.8
0.6
0.4
0.2
0.0
0 2 4 6 8 10 12 14 16 18 20
Months
Number at Risk
8 8 8 7 5 2 2 2 2 2 0

Figure 4: Kaplan-Meier Curve of Overall Survival (OS)

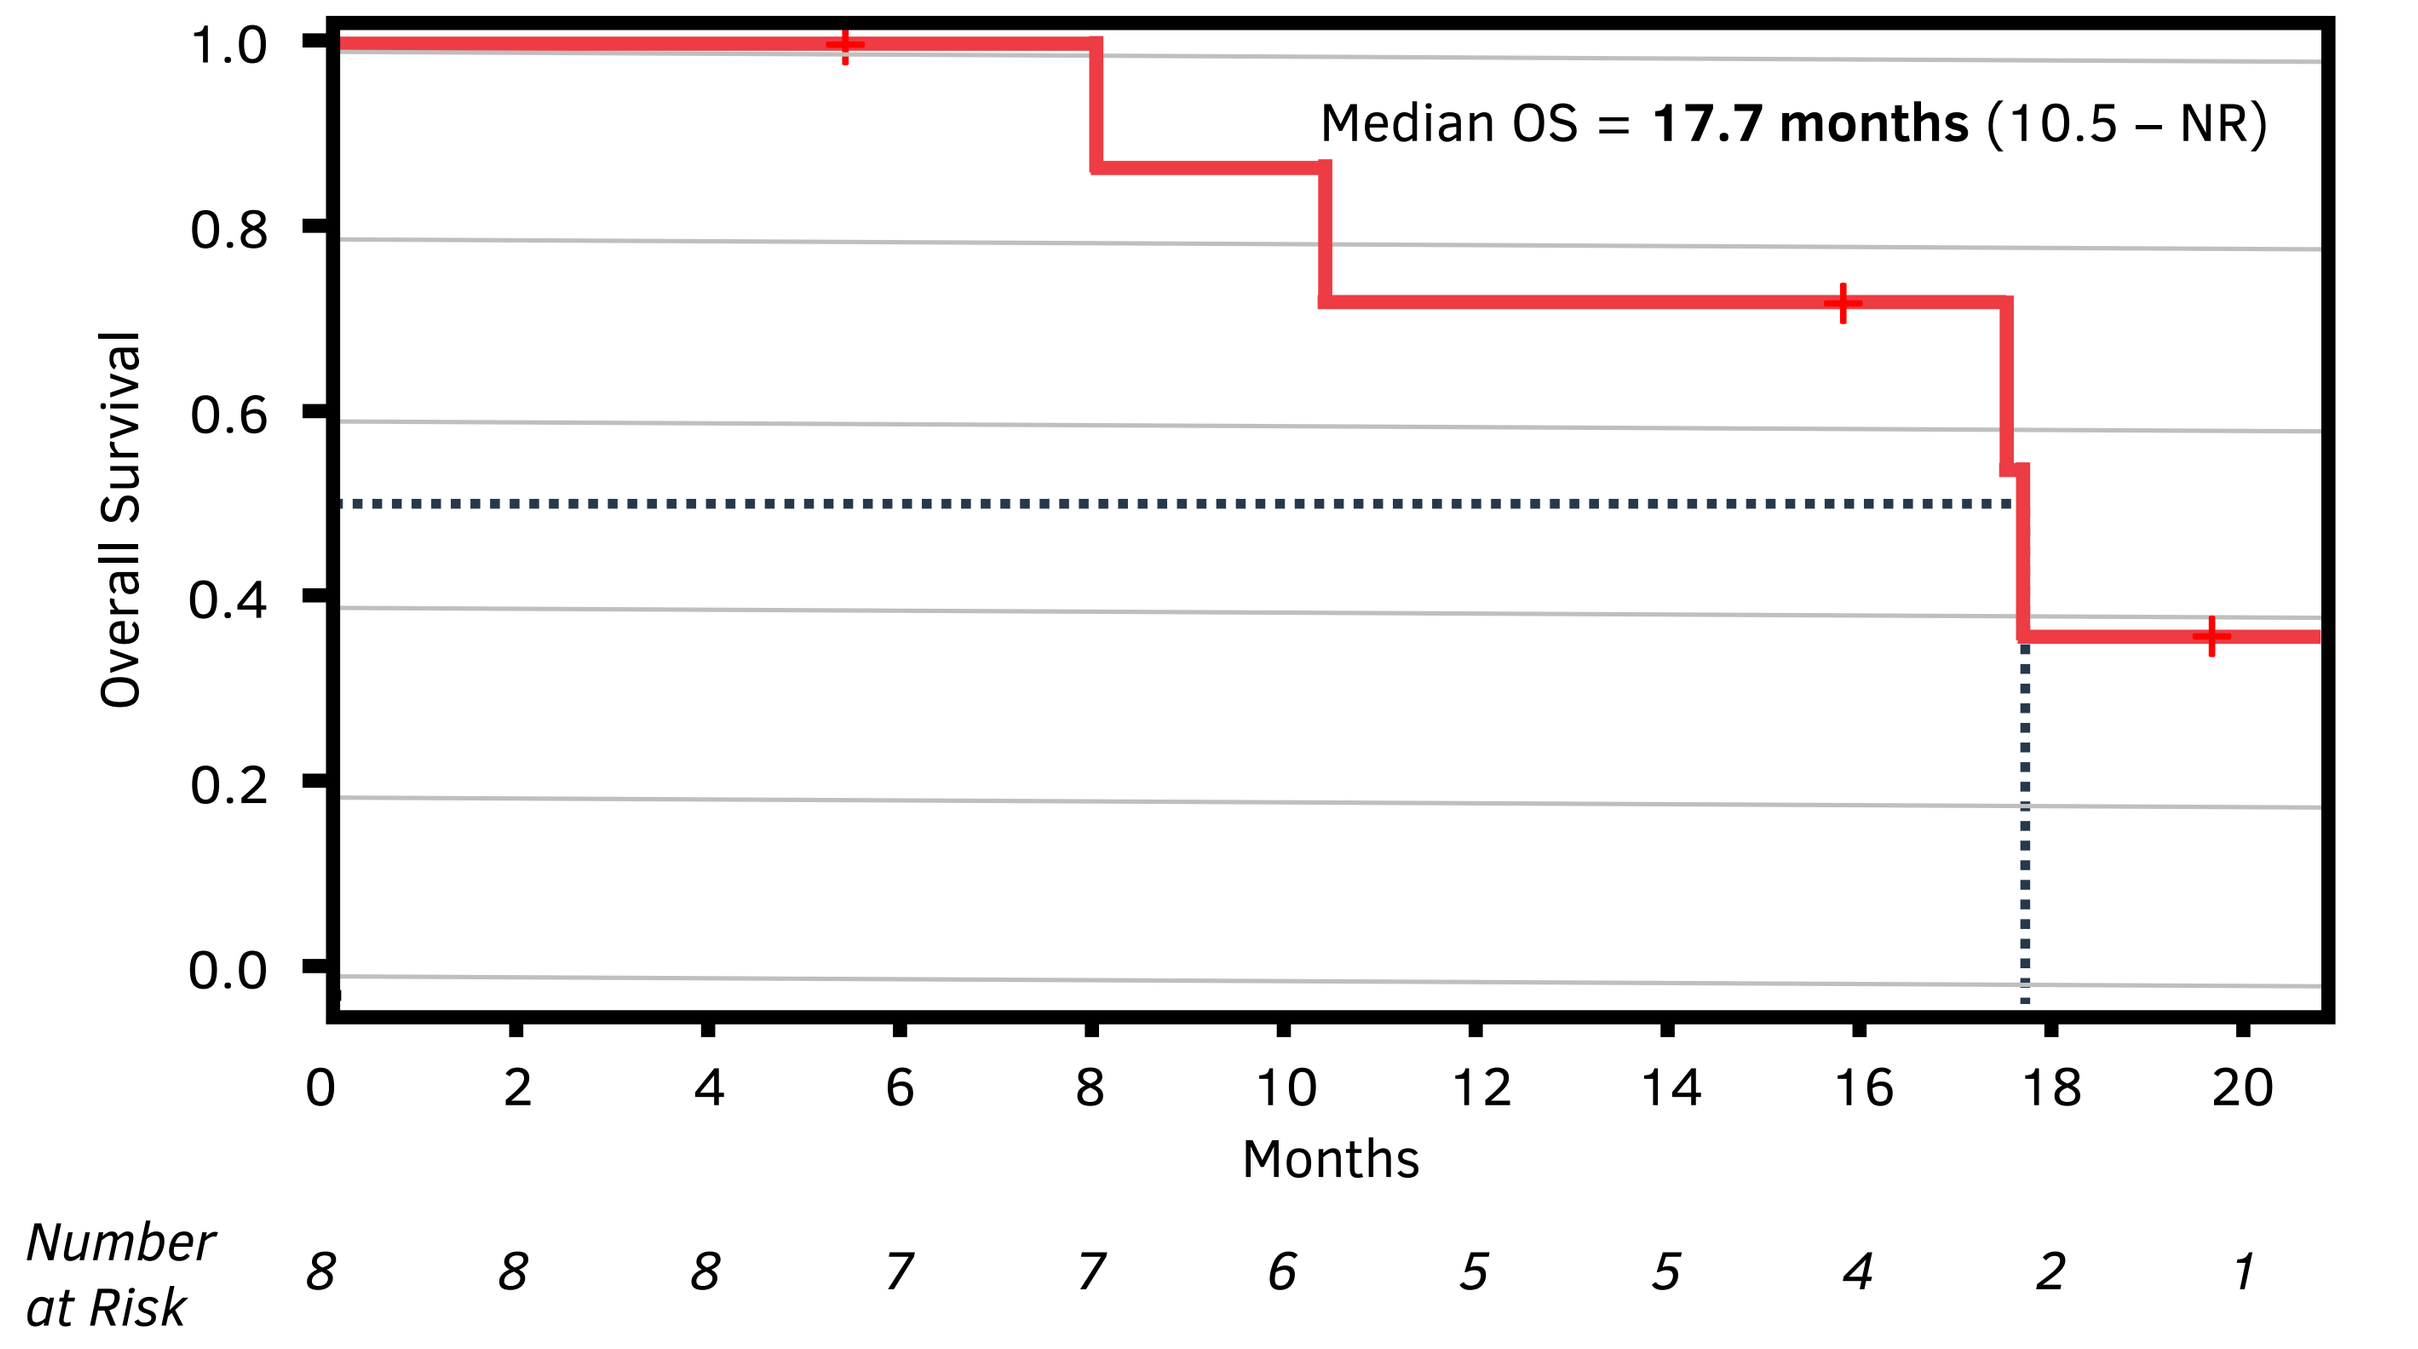
Median OS = 17.7 months (10.5 – NR)
Overall Survival
1.0
0.8
0.6
0.4
0.2
0.0
0 2 4 6 8 10 12 14 16 18 20
Months
Number at Risk
8 8 8 7 7 6 5 5 4 2 1

Analysis Cut-Off: 29 February 2020

DISCUSSION

Emerging Conclusions

- An **MTD of 60mg** has been determined for the newly-diagnosed population (versus 45mg for recurrent patients in the prior phase I study)
- Toxicities seem broadly **consistent with prior clinical experience** and with other PI3K-targeting agents
- GDC-0084 shows encouraging indications of potential clinical activity which compare favourably against historical controls
- Stage 2 (expansion cohort) is fully enrolled and in follow-up

Directions for Future Research

- A pivotal study in glioblastoma is planned to commence recruitment in 2H CY2020
- Phase I studies are also underway in DIPG and DMGs (NCT03696355), and in brain metastases in combination with radiotherapy (NCT04192981), and phase II studies are in progress in brain metastases (NCT03994796), and in HER2+ breast cancer brain metastases (NCT03765983)
- Potential use of the drug in recurrent glioblastoma and in methylated newly-diagnosed patients is under consideration

REFERENCES

1. Heffron TP *et al.* ACS Med Chem Lett. 2016; 7(4): 351-356.
2. Salphati L *et al*. Drug Metab Dispos. 2016; 44(12): 1881-1889.
3. Brennan CW *et al.* Cell 2013; 155(2): 462-477.
4. Wen PY *et al.* Clin Cancer Res. 2020; 26(8): 1820-1828

ACKNOWLEDGEMENTS

The authors wish to thank the patients and their families for participating in this study.

FUNDED BY KAZIA THERAPEUTICS LIMITED, AUSTRALIA

CONTACT – INFO@KAZIATHERAPEUTICS.COM

Scan me

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

American Society for Clinical Oncology (ASCO) – Annual Meeting 2020
Virtual – 29-31 May 2020

KAZIA THERAPEUTICS